A Randomized, Double-Blind, Placebo-Controlled Study of Topiramate in the Treatment of Tourette Syndrome

Joseph Jankovic, MD[1], Joohi Jimenez-Shahed, MD[1],
Lawrence W. Brown, MD[2]
[1]Baylor College of Medicine, Houston, TX;
[2]Children's Hospital of Philadelphia, PA

ABSTRACT

Objective: To investigate the effects of topiramate on Tourette syndrome. Background: Dopamine receptor blocking drugs have been traditionally used to control tics in patients with TS, but these neuroleptics are associated with potentially limiting side effects. Methods: This is a randomized, double-blind, placebo-controlled, parallel group study. To be included in the study, subjects required a DSM-IV diagnosis of TS, were 7-65 years of age, had moderate to severe symptoms (Yale Global Tic Severity Scale or YGTSS >19), were markedly impaired as determined by the Clinical Global Impression (CGI) scale severity score of >4, and were taking no more than one drug each for tics or TS co-morbidities. Results: There were 29 patients (26 males), mean age 16.5 ± 9.89, randomized and 20 (69%) completed the double-blind phase of the study. The primary endpoint was Total Tic Score, which improved by 14.29 ± 10.47 points from baseline to visit 5 (day 70) with topiramate (mean dose 118 mg) compared to 5.00 ± 9.88 point change in the placebo group (p = 0.0259). There were statistically significant improvements also in the other components of the YGTSS as well as improvements in various secondary measures, including the CGI and premonitory urge CGI. No differences were observed in the frequency of adverse events between the two treatment groups. Conclusion: This double-blind, placebo-controlled trial provides evidence that topiramate may have utility in the treatment of moderately severe TS.

BACKGROUND

Tourette syndrome (TS) is a complex neurological disorder characterized by multiple motor and phonic tics and a wide range of behavioral problems, including attention deficit with or without hyperactivity, obsessive-compulsive behavior, and impulse control problems (Jankovic, 2001). The pharmacological treatment of TS usually consists of antidopaminergic drugs and a wide array of other drugs, but the frequent occurrence of side effects limits their usefulness. There is, therefore, an unmet need for safer and more effective pharmacological treatments of tics and TS behavioral co-morbidities.

Topiramate is a broad-spectrum antiepileptic drug used as adjunctive and monotherapy in a wide variety of seizures. It has also been reported to be effective the management of headache, mood and behavioral disorders, pain, and tremors (Ondo et al, 2006). The exact mechanism of action of topiramate is unknown, but the drug has been found to enhance γ-aminobutyric acid subtype A (GABAA)-receptor mediated chloride flux and thus may potentiate GABA-mediated inhibition (Lyseng-Williamson et al, 2007). Topiramate also blocks the AMPA/kainate subtype of glutamate receptors without affecting the N-methyl-D-aspartate (NMDA) subtype. In humans, topiramate has been found to increase cerebral GABA, homocarnosine, and pyrrolidinone concentrations; it also acts as a weak carbonic anhydrase inhibitor. Topiramate is rapidly absorbed, with peak serum concentrations observed 1-4 hours after ingestion, readily enters the brain, and has over 80% bioavailability. Topiramate has clear advantages over neuroleptic medications in that it carries no risk of tardive dyskinesia or excessive weight gain.

METHODS

This is a multicenter, randomized, double-blind, placebo-controlled, parallel group study designed to investigate the change in severity of TS tic symptoms following treatment with topiramate when compared to placebo. In order to meet the inclusion criteria the subjects must have a DSM-IV diagnosis of TS for at least 3 months, be between 7 and 65 years of age, and weigh over 25kg. Subjects must have a minimum Yale Global Tic Severity Scale (YGTSS) rating scale of >19 and a Clinical Global Impression (CGI) scale severity score of >4 at Visit 1 (Day -10 through Day -7) and Visit 2 (Day 1). Subjects taking more than one agent for the treatment of tics or more than one agent for the treatment of co-morbid behavioral symptoms were excluded.

The selected subjects underwent a screening/washout visit (up to 30 days prior to randomization, 90 days if subject had been treated with botulinum toxin), before being randomized to receive either topiramate or placebo in a 1:1 ratio according to a computer-generated code. Subject and investigator were blinded to treatment assignment. During the titration phase, the study medication, consisting of 25 mg of topiramate or a matching identically appearing placebo tablet(s), was gradually increased over 6 weeks up to 200 mg/day, depending on tolerance. All subjects had to reach a minimum dose of 50 mg/day in order for them to proceed to the 4-week maintenance phase, followed by 12-day taper phase.

The following evaluations were performed on visit 2 (day 1) and visit 5 (day 70): YGTSS, CGI, Yale-Brown Obsessive-Compulsive Scale (Y-BOCS) or Children's Yale-Brown Obsessive-Compulsive Scale (CY-BOCS) (subjects <17 years), and Conners' Parent Rating Scale – Revised (L) (CPRS-R:L) (parent of subjects <17 years) or Conners' Adult A-D/HD Rating Scale – Self-Report: Long Version (CAARS-S:L) (subjects >18 years). Some of the instruments were also administered during visit 3 (day 28) and visit 4 (day 56), and the subjects were contacted by telephone weekly in between the visits. In addition to these assessments, vital signs and a variety of laboratory tests were performed as part of safety monitoring.

The efficacy analyses was based on the intent-to-treat population that included all subjects who had taken at least one dose of study medication and had at least one post baseline efficacy evaluation. The primary efficacy variable was the change of the Total Tic Score of the YGTSS at the last visit compared to day 1. Changes of individual components of YGTSS were compared using analysis of covariance with the baseline score at day 1 as a covariate. Secondary efficacy variables included CGI, premonitory urge CGI, Y-BOCS/CY-BOCS, and CPRS-R:L/CAARS-S:L.

Treatment groups were compared based on the incidence of adverse events and of adverse events of special interest based on Fisher's Exact Test. All statistical tests were two-sided at the 5% level of significance. Safety assessment was performed on the safety population, defined as all randomized subjects who received at least one dose of study drug. In the case of early withdrawal, data was obtained using the last observation carried forward (LOCF) method.

RESULTS

• There were 29 patients (26 males), mean age 16.5 ± 9.89, randomized to topiramate (N=15) or placebo (N=14) arms (Table 1). Twenty (69%) patients completed the double-blind phase of the study.

Table 1

Demographics and Baseline Characteristics

Variable	Topiramate (N=15)	Placebo (N=14)	Total (N=29)
N Randomized	15	14	29
N Completed*	12	8	20
Discontinued because of lack of efficacy	1	4	5
Discontinued because of limiting AEs	1	1	2
Lost to follow-up	1	1	2
Mean Age (years)**	18.8 ± 10.93	14.1 ± 8.35	16.5 ± 9.89
% Males	93	86	90
Mean Weight (kg) **	66.8 ± 22.71	60.4 ± 28.08	63.7 ± 25.19
Mean Height (cm) **	161.7 ± 18.14	155.9 ± 18.34	158.9 ± 18.13

Note: this table is based on the randomized population.

* Completed the double-blind phase (Visit 5)

** Baseline mean ± standard deviation

AEs = Adverse events

• In addition to TTS, there were also statistically significant improvements in the other components of the YGTSS, including the Total Motor Tic Score (TMTS), Total Phonic Tic Score (TPTS), and the Global Severity Score (GSS) (Table 2).

Table 2
Efficacy and Results

Variable	Topiramate (N=14)	Placebo (N=13)	P value
Mean TTS (baseline)	26.64 ± 8.78	28.77 ± 7.53	
Mean TTS (visit 5)	12.36 ± 12.04	23.10 ± 8.99	
Mean change from baseline TTS (visit 5)*	-14.29 ± 10.47	-5.00 ± 9.88	0.0259
Mean TMTS (baseline)	13.93 ± 4.70	14.23 ± 4.21	
Mean TMTS (visit 5)	8.14 ± 5.95	12.80 ± 6.63	
Mean change from baseline TMTS (visit 5)	-5.79 ± 5.67	-1.30 ± 4.35	0.0493
Mean TPTS (baseline)	12.71 ± 5.36	14.54 ± 5.38	
Mean TPTS (visit 5)	4.21 ± 6.72	10.30 ± 5.93	
Mean change from baseline TPTS (visit 5)	-8.50 ± 6.10	-3.70 ± 8.41	0.0435
Mean GSS (baseline)	57.36 ± 20.04	58.00 ± 18.85	
Mean GSS (visit 5)	20.21 ± 24.96	50.10 ± 18.08	
Mean change from baseline GSS (visit 5)	-37.14 ± 24.73	-8.00 ± 22.48	0.0030

Note: this table is based on the ITT population

* A component of the Yale Global Tic Severity Scale (YGTSS), the change from baseline in Total Tic Score (TTS) at visit 5 (day 70) is the pre-defined primary endpoint

TMTS = Total Motor Tic Score

TPTS = Total Phonic Tic Score

RESULTS - continued

• The Total Tic Score (TTS), the primary endpoint, improved by 14.29 ± 10.47 points from baseline to visit 5 (day 70) with topiramate (mean dose 118 mg) compared to 5.00 ± 9.88 point change in the placebo group (p = 0.0259) (Figure 1).

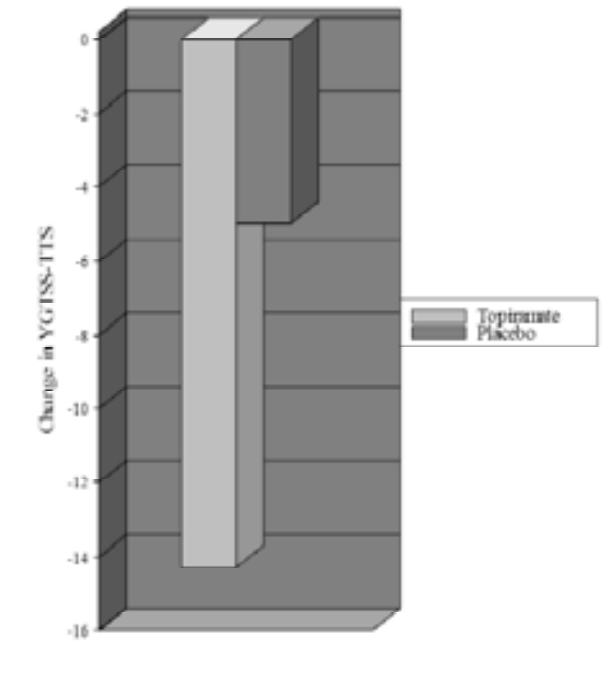

Change in YGTSS-TTS
0
-2
-4
-6
-8
-10
-12
-14
-16
Topiramate
Placebo

Figure 1 TTS in response to topiramate vs placebo

• The CGI and the premonitory urge CGI improved. No clinically significant differences were observed in the secondary measures, the frequency of adverse events or in the laboratory values between the two treatment groups (Table 3).

Table 3
Adverse Events

Variable	Topiramate (N=15)	Placebo (N=14)
≥ 1 AE	11 (73%)	13 (93%)
Headache	3 (20%)	3 (21%)
Diarrhea	3 (20%)	1 (7%)
Abdominal pain	2 (13%)	2 (14%)
Drowsiness/Hypersomnia	2 (13%)	2 (14%
Cognitive slowing	1 (7%)	0
Kidney stone	1 (7%)	0
Mean weight change from baseline to visit 5 (day 70)(kg)	-2.1	1.9

Note: this table is based on the safety population.

CONCLUSION

• Topiramate, as monotherapy or as an add-on to established therapy, is an important addition to the armamentarium in the treatment of TS.

REFERENCES

• Jankovic J: Tourette's syndrome. N Engl J Med 2001;345:1184-92.
• Lyseng-Williamson KA, Yang LP. Topiramate: a review of its use in the treatment of epilepsy. Drugs 2007;67:2231-56.
• Ondo WG, Jankovic J, Connor GS, et al; Topiramate Essential Tremor Study Investigators. Topiramate in essential tremor: a double-blind, placebo-controlled trial. Neurology 2006;66:672-7.

AKNOWLEDGEMENTS

The investigator-initiated study was supported by a grant from Ortho McNeil Janssen Scientific Affairs, L.L.C. We also thank P. Weisleder, MD and F. Leigh, MD (Duke University), and T. Nelson, MD (Carolina Neurological Clinic); C. Hunter, RN and E. Jimenez, MEd (Baylor College of Medicine); and Y. Collins, RN and Susan Melamed, PNP (Children's Hospital of Philadelphia), for their assistance.