Eliminating health disparities in cancer clinical trials: The EDICT Project

Barbara C. Pence[1], Daniel Bustillos, Armin Weinberg[2]

[1]Texas Tech University Health Sciences Center, Lubbock, TX, [2]Baylor College of Medicine, Houston, TX

Abstract:

In 1993 the NIH Revitalization Act was enacted, and it requires applicants for federal research funding to provide a strategy for inclusion of people of diverse racial and ethnic origin and women into clinical trials. In spite of the Revitalization Act, current research indicates that underrepresented groups are still not adequately represented in cancer clinical trials. According to the Coalition of Cancer Cooperative Groups, 89% of participants enrolled in NCI clinical trials between January 2003 and June 2005 were white. In addition to the clear emphasis of the Belmont Report on equity and justice in both burdens and benefits of clinical research, social justice has not been achieved when it comes to inclusion of the underrepresented in cancer clinical trials. Without adequate representation of these populations in clinical trials, researchers cannot learn about differences in safety and efficacy between groups and cannot ensure generalization of scientific results. This also contributes to health disparities by keeping the newest techniques and drugs out of the reach of the disadvantaged groups. This concern for the overwhelming underrepresentation of diverse groups in cancer clinical trials specifically, has led to the development of the EDICT (Eliminating Disparities in Clinical Trials) Project*, a joint project between Baylor College of Medicine and the Intercultural Cancer Council. The EDICT mission is to address problems and solutions related to improving the participation of minority and underserved populations in clinical trials. The underlying premise of the EDICT Project is the need for a change in how we design and oversee clinical research so that there is accountability and enforcement for equitable inclusion in clinical trials, and fostering research design which takes into account not only equitable participant accrual, but also differential plans for retention of underrepresented populations. One early focus of EDICT has been policy research, while ongoing EDICT field demonstration research will test the effect of various interventions on participant accrual and retention rates in clinical trials when applied to underrepresented populations, as compared to no intervention. It is anticipated that the outcomes of the EDICT Project will benefit all of the underserved, whether the reason for underrepresentation be race, ethnicity, elderly status or rural domicile.

*** Funded by Genentech, Inc.**

EDICT Project:
A 4-year collaboration between BCM, Intercultural Cancer Council.
Addresses problems and solutions related to improving participation of minority and underserved in clinical trials.
Funded by Genentech, Inc.
CREDO: The following beliefs guide our work together:
All individuals will have the opportunity and necessary support to participate voluntarily in clinical trials for which they are eligible.
Participants and researchers will understand and promote the benefits of diversity in clinical trials.
Results from clinical research will benefit the participant's communities and society at large.

Opportunity Teams:
OT1 **Allocation of Research Funding Proportionate to Case Fatality**
OT2 **Assuring Healthcare Coverage in Clinical Trials**
OT3 **Education and Training - Institutional and Professional**
OT4 **Education and Training - Public and Patients**
OT5 **Fostering Community Input and Involvement and Relationships**
OT6 **Patient/Participant Navigation**
OT7 **Pharmaceutical Partnerships**
OT8 **Publication-Related Policies**
OT9 **Regulatory Oversight and Enforcement**

Policy Research Objectives:
assess policy issues related to underrepresented populations participation in clinical trials,
organize and conduct a National Policy Development Summit Meeting, and
conduct dissemination activities involving policy education and advocacy relating to minority/underrepresented participation in clinical trials.

Website:

http://www.bcm.edu/edict/home.html

Call for Culture Change:
What was the original impetus for the current legislative and regulatory environment for clinical research oversight?
Most immediate concern: safety and protection from exploitation.
Belmont Report also has a clear emphasis on equity and justice.
Today's oversight bodies are generally more concerned with participant safety
Some see justice and equity to be completely outside the purview of human subjects research oversight.
We propose that a significant "culture shift" is in order to rehabilitate the original intent and impetus of HSR oversight in this country.

What this might look like:
Accountability and Enforcement for equitable inclusion in clinical trials
Research design takes into account not only equitable participant accrual but also differential plans for retention of underrepresented populations.
Call for Culture Change
Inclusion policies cannot summarily exclude those with disabilities or co-morbidities (that don't significantly heighten the risk to participants or that don't confound the science).
CFR is amended to require IRB composition to be more representative of community in which the trial is to take place.
Consent forms and instructional material as well as interpretation services are made available in all of the area's major languages (Perhaps any language spoken by at least 5% of the area's population).

•Goal:

The Field Demonstration research arm of EDICT is designed to test the effect of various interventions on participant accrual and retention rates in cancer and asthma trials when applied to under-represented populations, as compared to no intervention.

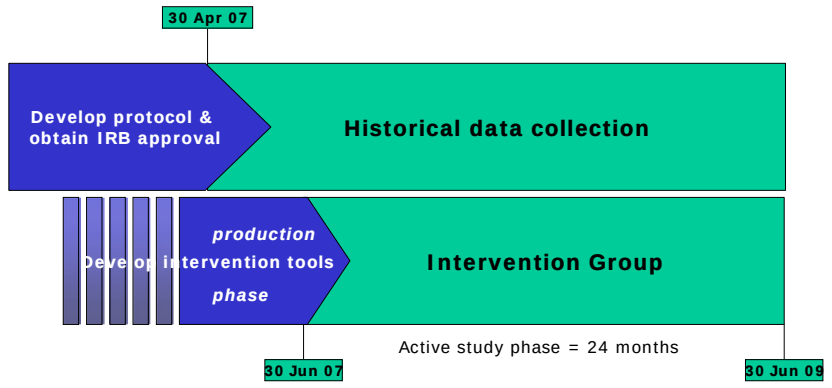
30 Apr 07
Develop protocol & obtain IRB approval
Historical data collection
Develop intervention tools production phase
Intervention Group
Active study phase = 24 months
30 Jun 07
30 Jun 09

CLAS-ACT & EDICT BackPack Projects

❖National Standards for Culturally and Linguistically Appropriate Services

….. and Clinical Trials – "CLAS-ACT"

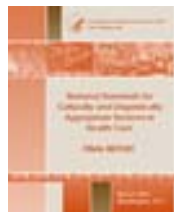

❖CLAS mandates are current Federal requirements for all recipients of Federal funds.
- CLAS guidelines are activities recommended by OMH for adoption as mandates by Federal, State, and National accrediting agencies.
- CLAS recommendations are suggested by OMH for voluntary adoption by health care organizations.

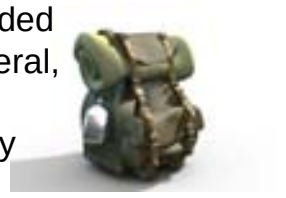

❖Identification of existing evidence-based and promising practices to support those addressing the elimination of disparities in clinical trials.
- Government, Private, Non-profits
- Exemplar sites
- Review process

❖DHHS OMH Partnerships to Eliminate Disparities

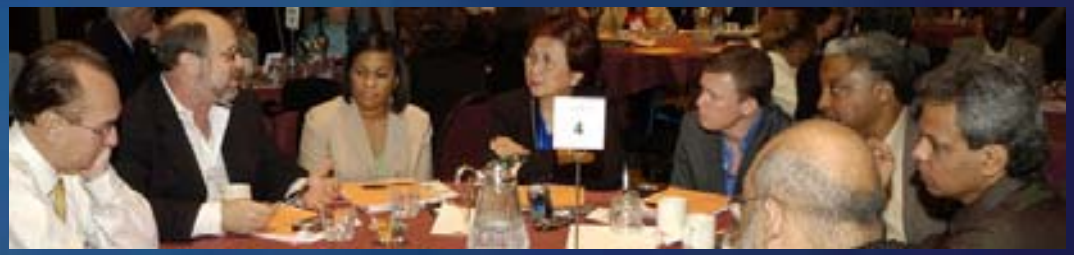

EDICT Eliminating Disparities in Clinical Trials